Therapeutic Promises of Cell-derived Vesicles (CDVs) as mRNA Delivery Platform

Jinhee Park, Kyeongeun Park, Seung Wook Oh, Hye Yeong Nam
BioDrone Research Institute, MDimune Inc., Seoul, Republic of Korea

Introduction

The tremendous success of mRNA vaccines against coronavirus disease 2019 (COVID-19) has renewed interest in mRNA therapeutics. However, efficient delivery of mRNA to target cells remains the major challenge in developing mRNA-based therapeutics. As reported, synthetic vehicles such as lipid nanoparticles and polymers commonly used for mRNA delivery still have concerns regarding immunogenicity and toxicity[1]. Here, we describe an alternative approach for mRNA delivery using cell-derived vesicles (CDVs) to overcome safety issues for mRNA therapeutics. The CDVs are obtained from diverse human cell sources by applying MDimune's proprietary extrusion technology. We have previously demonstrated that CDVs can serve as effective drug carriers with biocompatibility, excellent cellular uptake efficiency, and enhanced tissue penetration capability[2]. To verify the potential of CDVs as mRNA delivery carriers, we developed a methodology to deliver mRNAs using complexation of mRNA with CDVs via cationic reagent.

Characterization of mRNA-loaded CDVs

mRNAs were loaded onto the CDVs using ionizable cationic lipid, 8-[(2-hydroxyethyl)[6-oxo-6-(undecyloxy)hexyl]amino]-octanoic acid, 1-octylnonyl ester (SM-102), as shown in the schematic diagram (Fig. 1A). The mRNA-loaded CDVs were characterized by consistent size (< 300 nm) with a polydispersity index (PDI) of less than 0.3 and efficient mRNA loading (> 90%). Furthermore, the size and morphology of CDVs and mRNA-loaded CDVs were observed by cryo-TEM. The images showed that CDVs were detected as round particles with sizes ranging from 70-150 nm and the mRNA-loaded CDVs were 100-200 nm upon mRNA loading. No aggregates of mRNA-loaded CDVs were observed. These results demonstrated that the CDVs could be loaded with exogenous mRNAs, suggesting a novel mRNA delivery platform using CDVs.

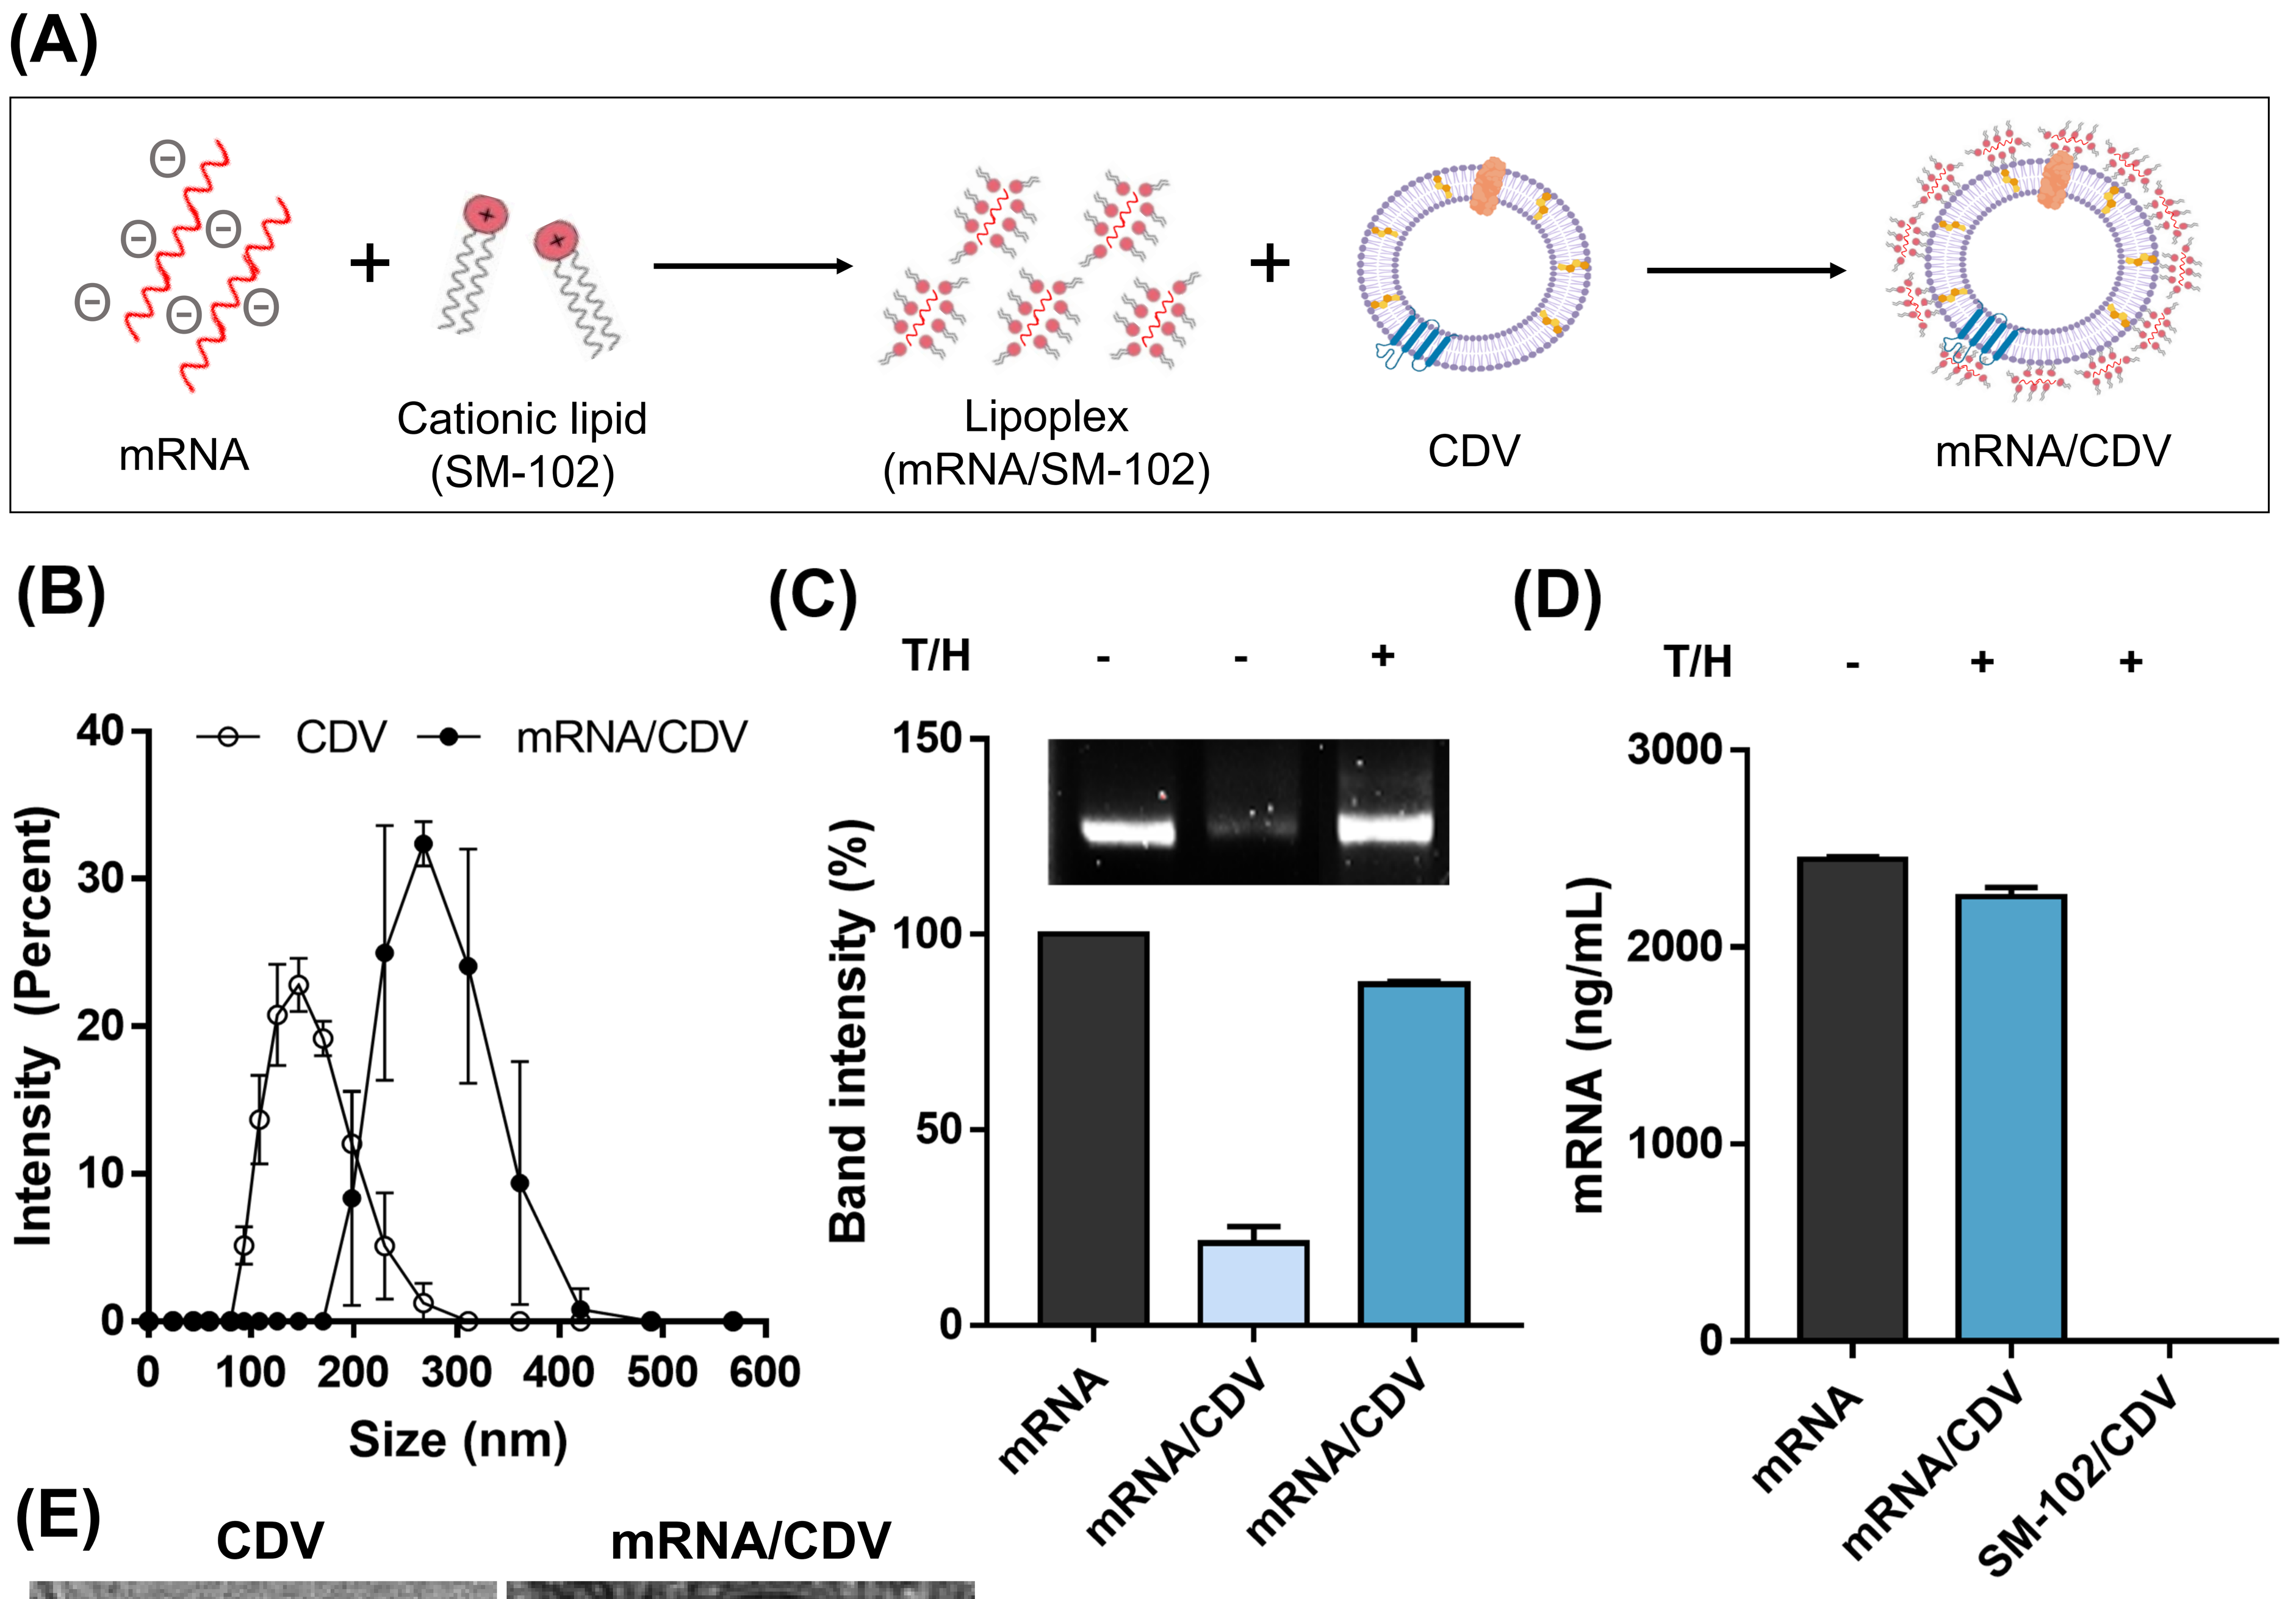
(A)
mRNA
Cationic lipid (SM-102)
Lipoplex (mRNA/SM-102)
CDV
mRNA/CDV
(B)
CDV
mRNA/CDV
Intensity (Percent)
Size (nm)
(C)
T/H
Band intensity (%)
mRNA
mRNA/CDV
mRNA/CDV
(D)
T/H
mRNA (ng/mL)
mRNA
mRNA/CDV
SM-102/CDV

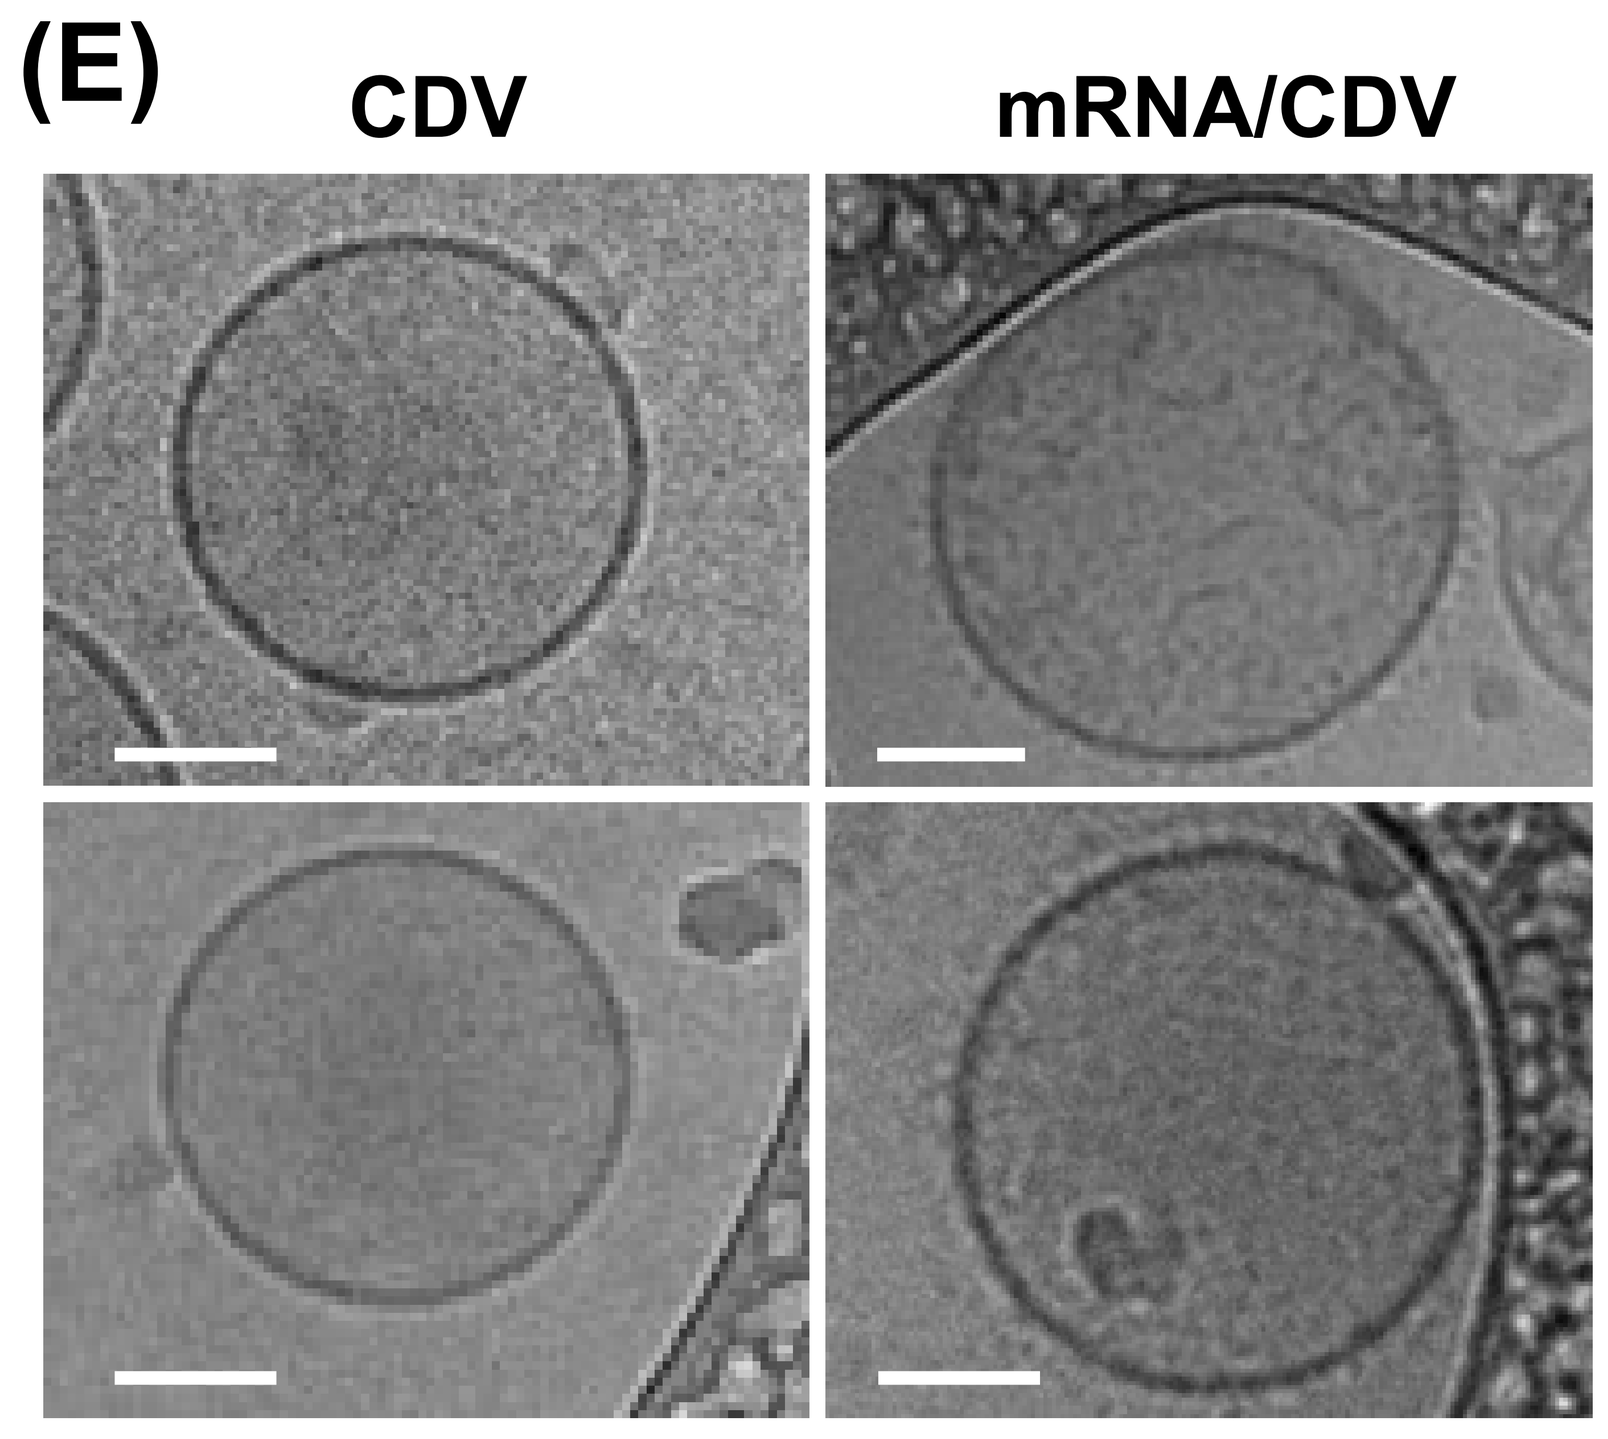
(E)
CDV
mRNA/CDV

Figure 1. (A) Schematic diagram shows mRNA loading method. (B) Size distribution profile and PDI of CDVs (open circles) and mRNA-loaded CDVs (closed circles) were determined by dynamic light scattering (DLS). (C-D) mRNA loading efficiency was investigated using agarose gel electrophoresis and fluorescence-based RNA quantification method (ribogreen assay). Triton X 100 (T) and heparin (H) were used to release mRNA from CDVs. (E) Representative cryo-TEM images of CDV and mRNA/CDV. Scale bar, 50 nm.

Single particle analysis

Distribution of loaded CDVs was analyzed using nanoparticle flow cytometry (nFCM) at the single particle level. CDVs were first labeled with CellTrace Far Red (CTFR) and then loaded with Alexa Fluor 488-conjugated mRNAs. The proportion of double-positive (CTFR and Alexa Fluor 488) showed that more than 90% of mRNAs were loaded onto CDVs, which was coherent with the analyses of agarose gel electrophoresis and RNA quantification.

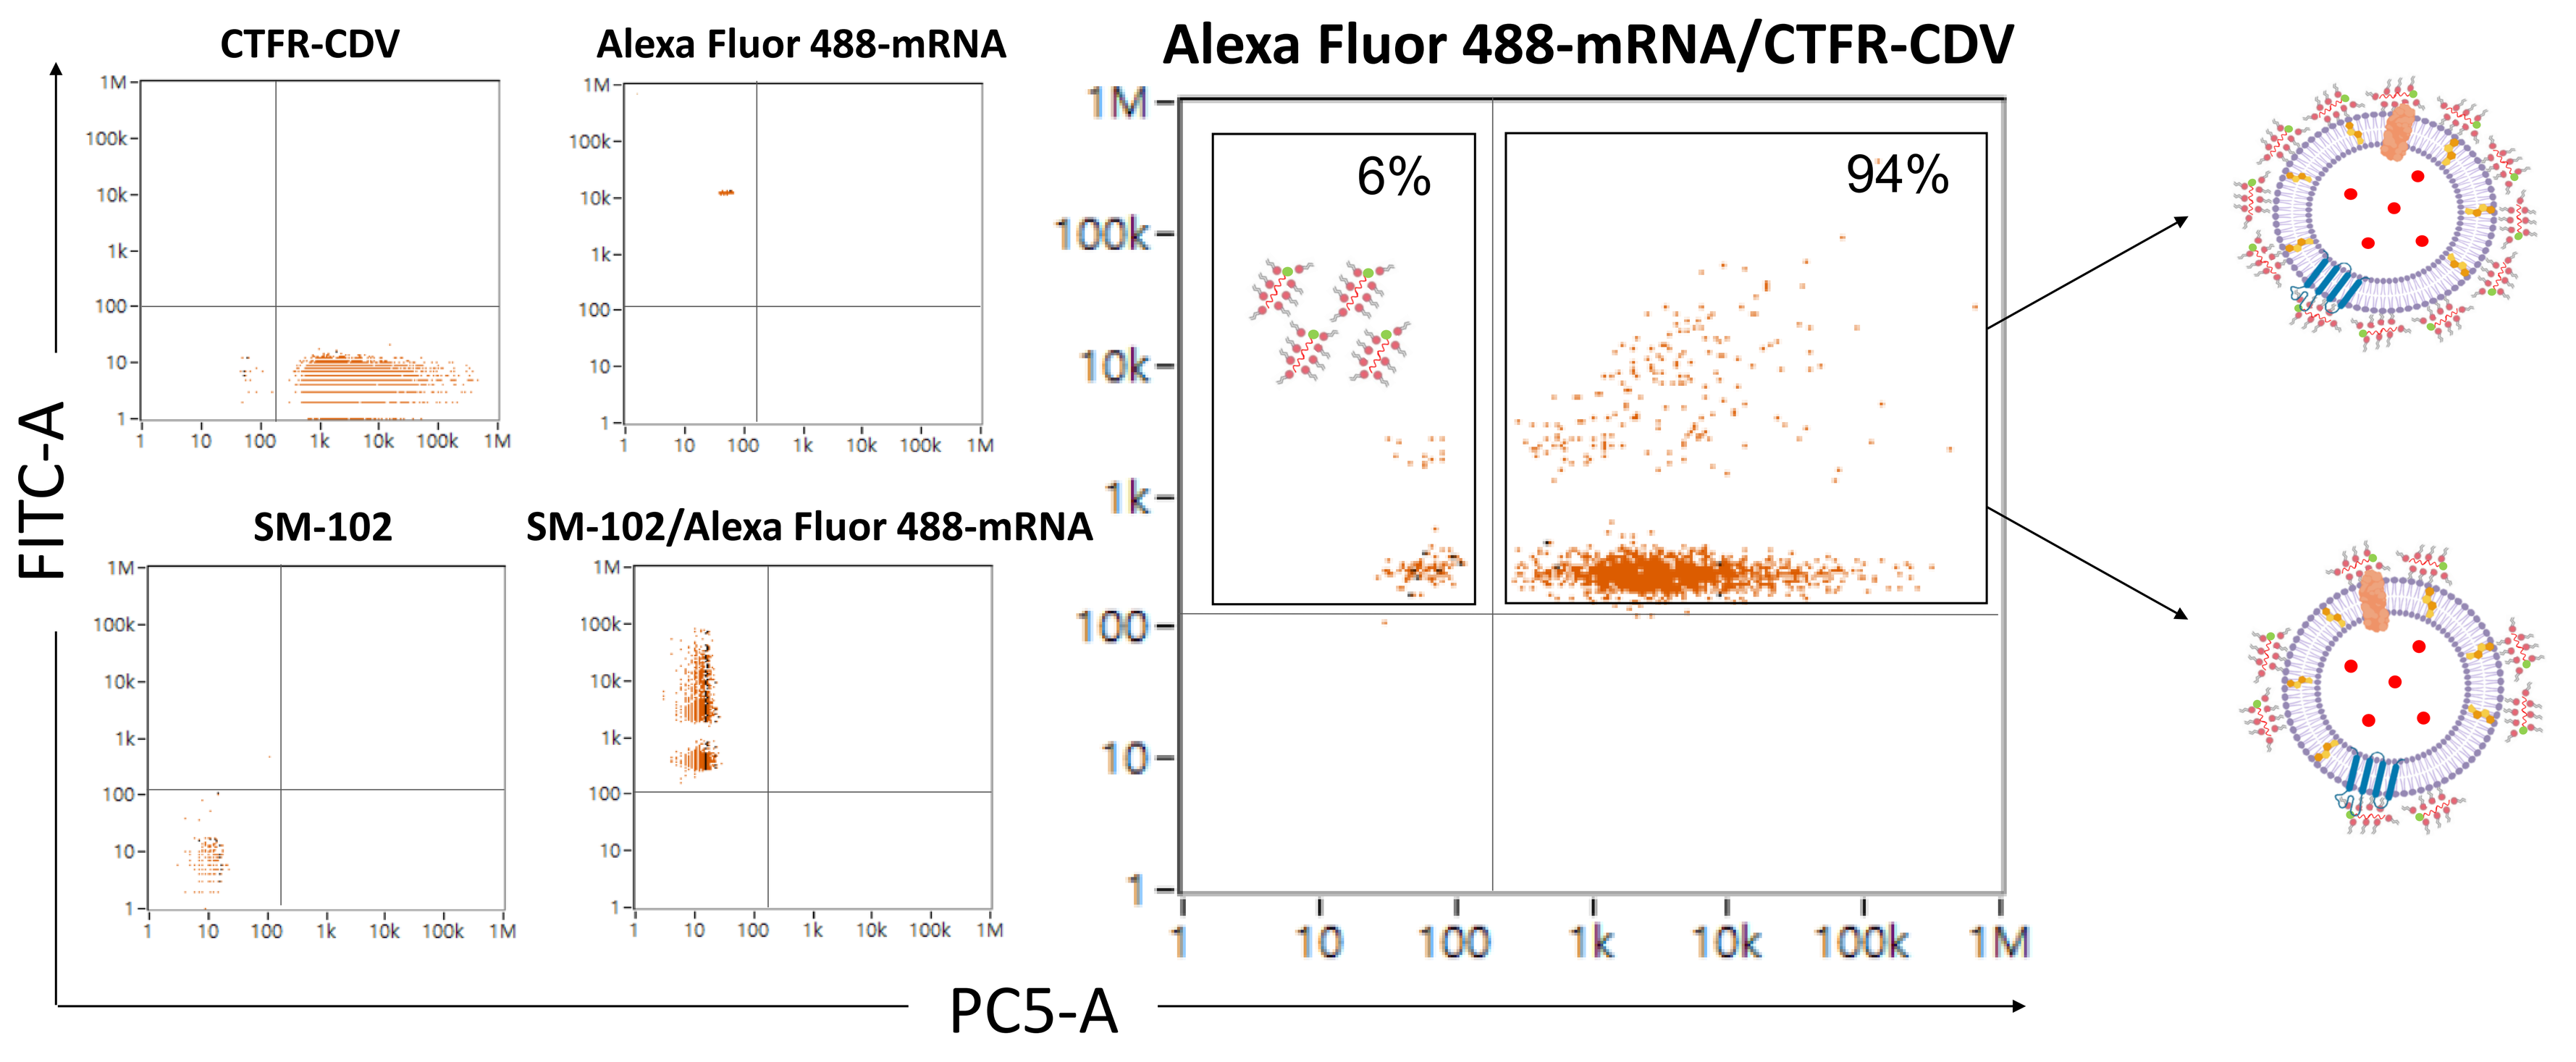
CTFR-CDV
Alexa Fluor 488-mRNA
SM-102
SM-102/Alexa Fluor 488-mRNA
Alexa Fluor 488-mRNA/CTFR-CDV
6%
94%
FITC-A
PC5-A

Figure 2. nFCM analysis of mRNA-loaded CDVs at the single particle level. Control groups consist of CTFR-labeled CDVs, Alexa Fluor 488-mRNAs, SM-102 and Alexa Fluor 488-mRNA /SM-102.

In vitro efficacy and toxicity

The efficacy and toxicity of the mRNA-loaded CDVs were evaluated at *in vitro* level. mRNA-loaded CDVs increased cellular uptake of mRNA into target cells compared to mRNA/SM-102 lipoplex alone. The mRNA-loaded CDVs also showed drastically enhanced protein expression in the treated cells, comparable to lipofectamine, with no significant cytotoxicity.

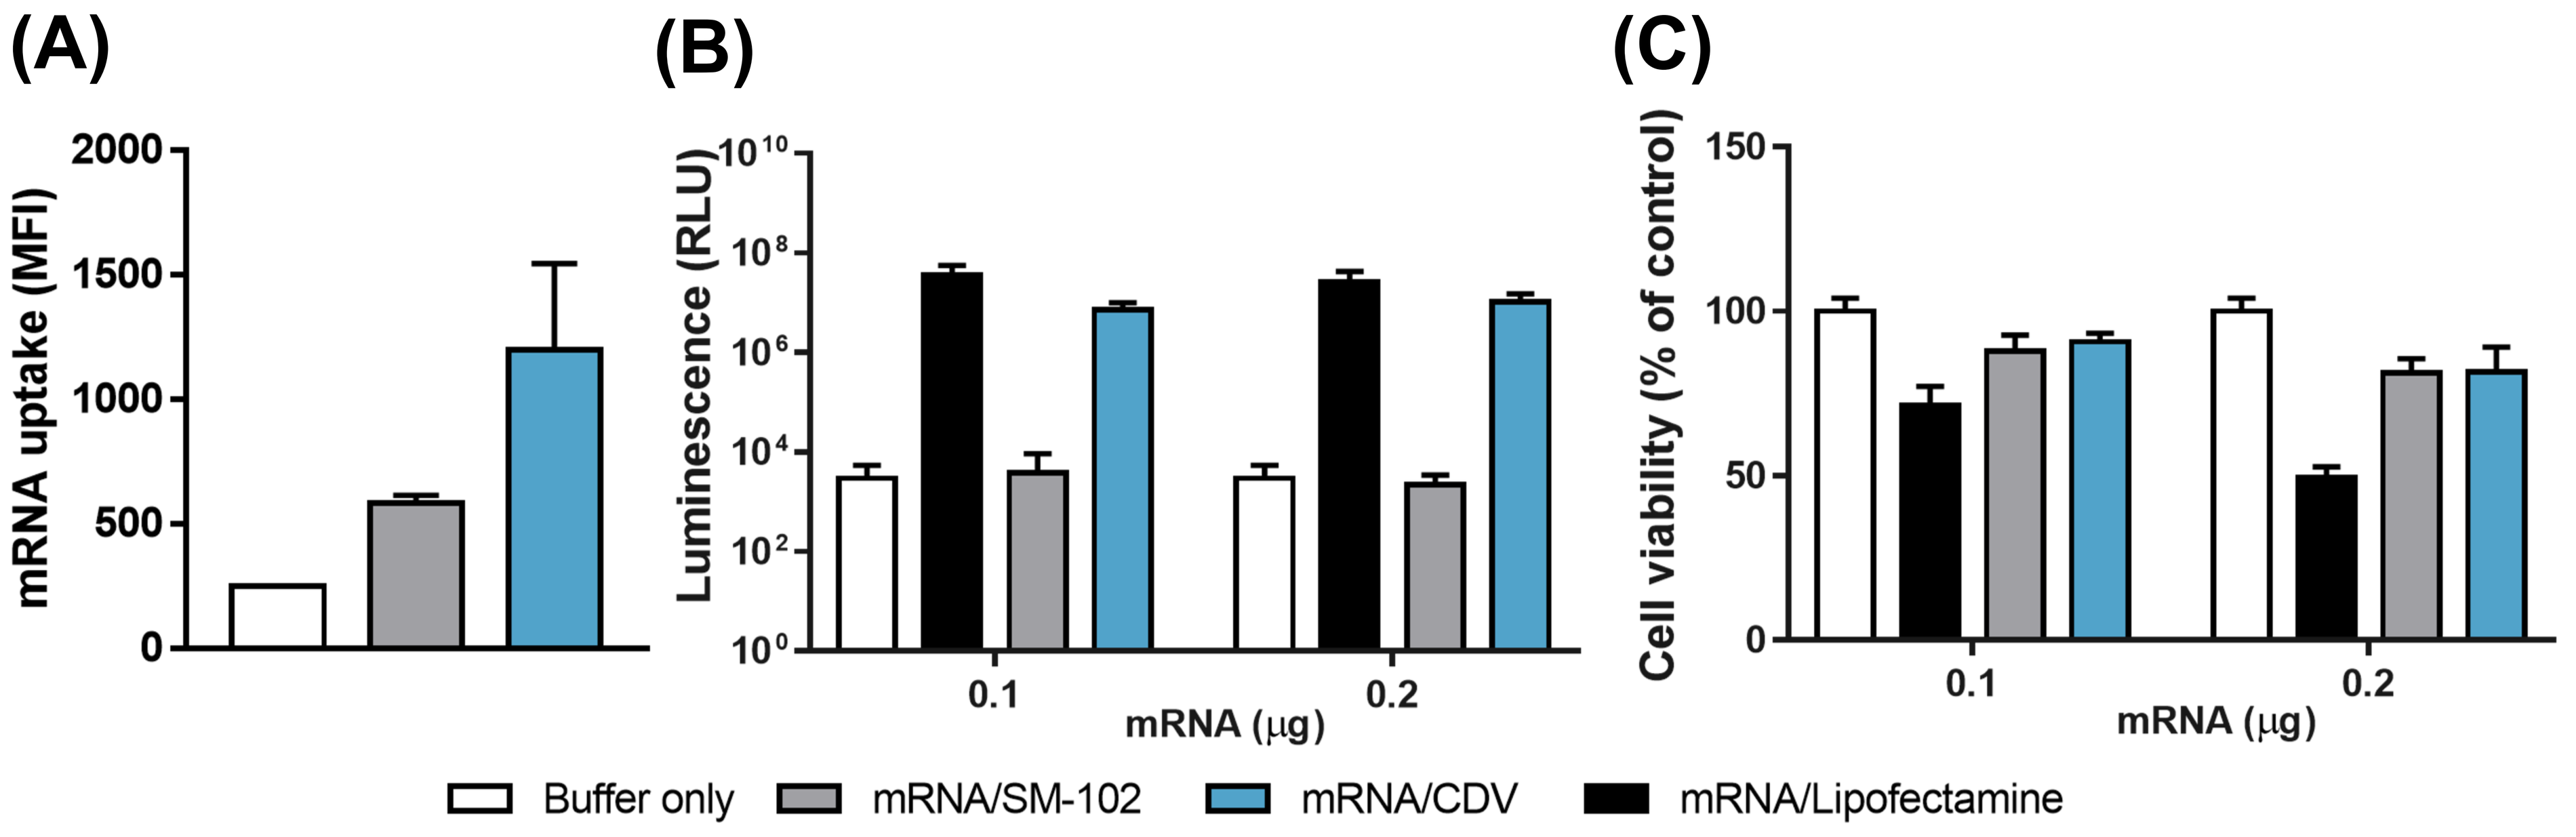
(A)
mRNA uptake (MFI)
(B)
Luminescence (RLU)
mRNA (μg)
(C)
Cell viability (% of control)
mRNA (μg)
Buffer only
mRNA/SM-102
mRNA/CDV
mRNA/Lipofectamine

Figure 3. *In vitro* evaluation of mRNA-loaded CDVs using HEK293 cells. (A) Cellular uptake of Alexa Fluor 488 mRNAs loaded onto CDVs was analyzed by flow cytometry. (B) Transfection efficiency of CDV loaded with Renilla luciferase mRNAs was evaluated by luciferase assay. (C) The cytotoxicity effect was assessed by CCK-8 assay after 24 h of incubation.

Storage stability

The stability of mRNA-loaded CDVs was assessed by the size, *in vitro* efficacy, and mRNA integrity. Loaded CDVs were stable up to 55 μg of mRNA/mL with 10-20% trehalose as an excipient. The stability of mRNA-loaded CDVs with different mRNA concentrations was maintained for up to 4 weeks at -80℃.

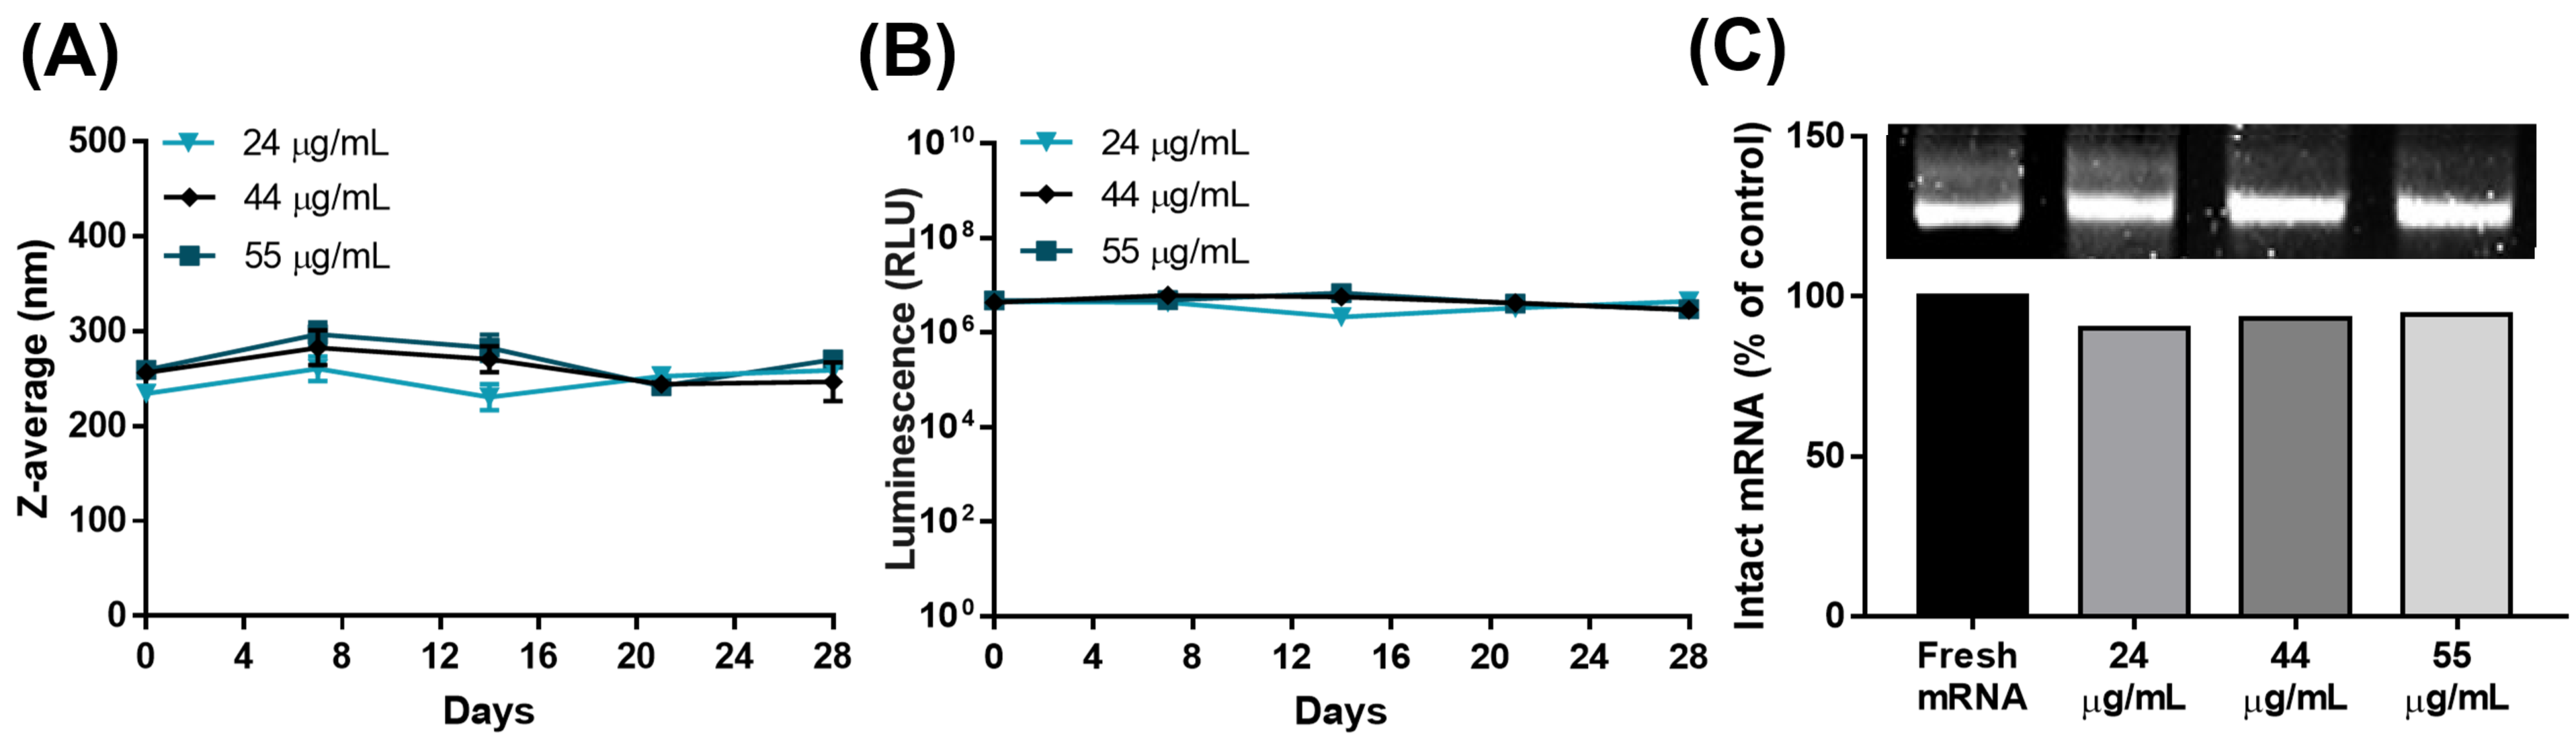
(A)
24 μg/mL
44 μg/mL
55 μg/mL
Z-average (nm)
Days
(B)
Luminescence (RLU)
Days
(C)
Intact mRNA (% of control)
Fresh mRNA
24 μg/mL
44 μg/mL
55 μg/mL

Figure 4. Long-term storage stability of mRNA-loaded CDVs at -80℃ (A) Size was measured by DLS. (B) *in vitro* efficacy was assessed by luciferase assay. (C) mRNA integrity was quantified by agarose gel electrophoresis.

Conclusion & Future Prospects

- We have demonstrated that CDVs facilitate the delivery of mRNA into target cells with low toxicity.
- The mRNA-loaded CDVs are stable at -80℃ for at least 4 weeks upon storage .
- We further aim to
 - investigate the biodistribution, efficacy, and toxicity of mRNA-loaded CDVs in an *in vivo* model.
 - load mRNAs encoding therapeutic proteins into CDVs and demonstrate the potential of CDVs as mRNA delivery carriers in various disease models.

[1] Cao, Y., and Gao, G. F. (2021) mRNA vaccines: A matter of delivery. EClinicalMedicine 32, 100746
[2] Lau H-C. (in press). GMP-compliant manufacturing of biologically active cell-derived vesicles produced by extrusion technology. Journal of Extracellular Biology.